P08-02

16th EFNS Congress of the European Federation of Neurological Societies , 8th-11th September 2012, Sweden

Cost effectiveness of Apomorphine infusion in the treatment of advanced Parkinson Disease in the UK

E. Walter[a], D. Mercsanits[a]

[a]IPF Institute for Pharmaeconomic Research, Vienna, Austria

Objectives

Parkinson Disease (PD) is the second commonest cause of neurological disability and affects 100-180 people per 100,000 of the population and increases with age. Continuous subcutaneous apomorphine represents an alternative treatment option of advanced PD with motor fluctuation. The purpose of this analysis was to estimate the cost-effectiveness of continuous s.c. Apomorphine infusion (CSAI) compared with Levodopa/carbidopa intestinal gel (LCIG), Deep-Brain-Stimulation (DBS) and Standard-of-care (SOC).

Methods

We developed a Markov-Model to simulate the long-term consequences, disease progression (Hoehn&Yahr-stages 3-5, percentage of waking-time in the OFF-state), complications and adverse-events. The model is adopted based on models which have been published for the UK and Sweden. Complications are different for the alternatives (e.g. pump problems in case of LCIG, temporary/permanent complications in case of DBS). Moderate and severe adverse-events (e.g. motor fluctuation, dyskinesia, nausea, dizziness, hallucination, skin problems, depression, anxiety) and death were included. Including 25 health-states, the model comprises moderate and severe health conditions. Probabilities derived from RCT and open-label studies; direct costs (2012 £) from published sources from the payer's perspective.

Fig. 1: Model Design

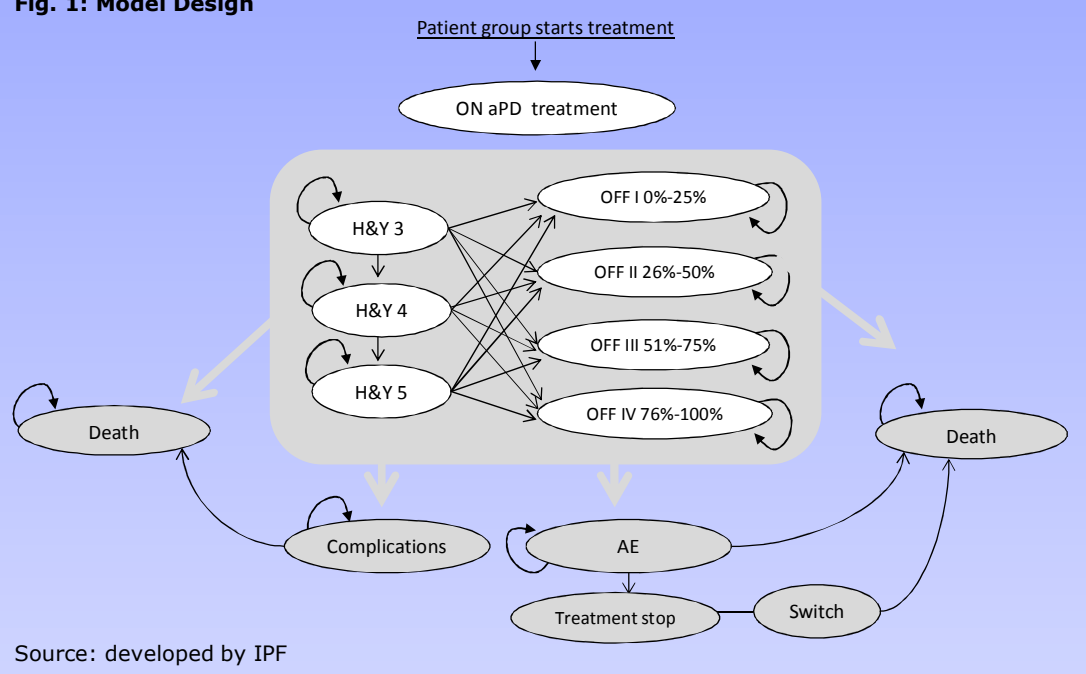
Patient group starts treatment
ON aPD treatment
H&Y 3
H&Y 4
H&Y 5
OFF I 0%-25%
OFF II 26%-50%
OFF III 51%-75%
OFF IV 76%-100%
Death
Death
Complications
AE
Treatment stop
Switch

Source: developed by IPF

The medical resource use and Utilities are based on literature research.

QALYs, life-years and costs are projected over a life-time horizon and discounted at 3.5%. Monte-Carlo-simulation accounted for uncertainty.

Cohort definition

The model cohort comprises PD patients aged 50 years or older with Hoehn&Yahr (H&Y) stages 3, 4 or 5 experiencing more than 50% of waking-time (14 hour) in the OFF state at treatment initiation. After treatment initiation with CSAI, LCIG or DBS an improvement in health states in the following cycle may arise; or patients continue SOC. During different therapeutic alternatives adverse events or complications may arise. Due to intolerable adverse events, patients have the opportunity to switch to an alternative treatment. Patients receiving first-line CSAI or LCIG switch to DBS and patients after a DBS either start or increase SOC medication.

Results

Over a life-time horizon, costs associated with CSAI amounts to 70,258.28 £ and 2.85 QALYs (6.28 LYs). Costs associated with LCIG are 117,121.48 £ and 3.06 QALYs (6.93 LYs). The cost-saving amounts to 46,863.20 £ per patient and the incremental-cost per QALY gained (ICER) was 223,051.88 £. Costs associated with DBS are 88,361.61 £ and 2.75 QALYs (6.38 LYs). CSAI dominates DBS. SOC associated total-cost are 68,082.92 £ and 2.62 QALYs (5.76 LYs). The ICER of CSAI versus SOC amounts to 9,296.41 £.

Tab. 1: Results of costs QALYs and LYs

Strategy	Costs C	Diff. ΔC	QALYs E	Diff. QALYs ΔE	ICER
CSAI	70,258.28 £		2.85		9,296.41 vs. SOC
LCIG	117,121.48 £	46,863.20 £	3.06	0.21	223,051.88 vs. CSAI
DBS	88,361.61 £	18,103.33 £	2.75	-0,10	(Dominated by CSAI)
SOC	68,082.92 £	-2,175.36 £	2.62	-0.23	
Strategy	**Costs C**	**Diff. ΔC**	**LYs E**	**Diff. LYs ΔE**	**C per LYs gained**
CSAI	70,258.28 £		6.28		4,225.64 vs. SOC
LCIG	117,121.48 £	46,863.20 £	6.93	0.65	72,075.05 vs. CSAI
DBS	88,361.61 £	18,103.33 £	6.38	0.11	170,144.08 vs. CSAI
SOC	68,082.92 £	-2,175.36 £	5.76	-0.51	

Source: IPF own calculations

ICER = incremental cost effectiveness ratio

Institute for Pharmaeconomic IPF Research

CONTACT:
Dr. Evelyn Walter
IPF Institute for Pharmaeconomic Research
Wolfengasse 4/7
1010 Vienna, Austria
Tel +43-1-5132007-13
Fax +43-1-5132007-15
Email: e.walter@ipf-ac.at
Web: www.ipf-ac.at

Fig. 2: Distribution of patients according to OFF time per 6 month

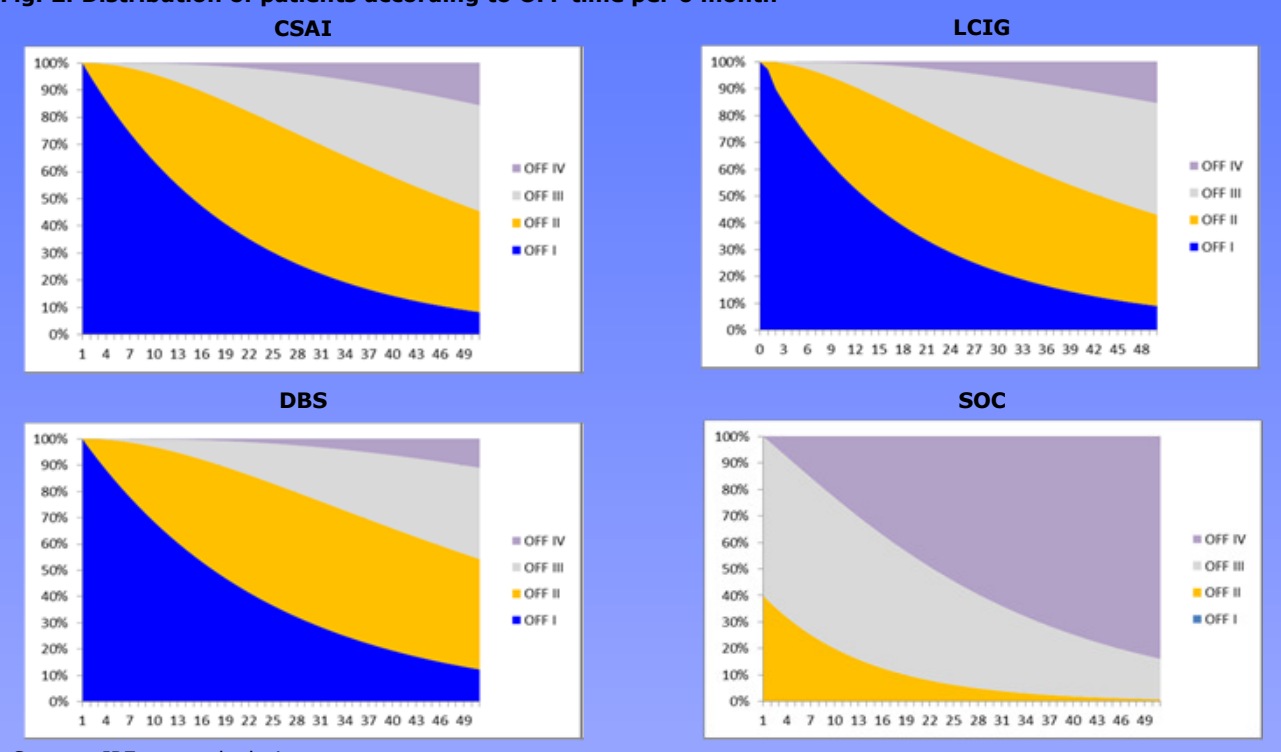
CSAI
LCIG
DBS
SOC
OFF IV
OFF III
OFF II
OFF I

Source: IPF own calculations

CSAI, LCIG and DBS patients spent less time in higher OFF states compared to patients maintained on SOC. Comparison of the graphs illustrates the better prognosis for patients treated with CSAI, who spent 69% of their remaining life-time in OFF I stage, 24% in OFF II, 6% of time in OFF III and 1% in OFF IV. Patients treated with LCIG spent 76% of their life-time after treatment start in OFF I, 20% in OFF II, 3% in OFF III and 1% in OFF IV. DBS shows the following effect: patients spent 78% of time in OFF I state, 19% in OFF II, 2.5% in OFF III and 0.5% in OFF IV.
In comparison to that, patients remaining on SOC therapy spend 27% of time in OFF II, 57% in OFF III and 16% in OFF IV stage.

Fig. 3a: Probabilistic Sensitivity Analysis **3b: Acceptability curves**

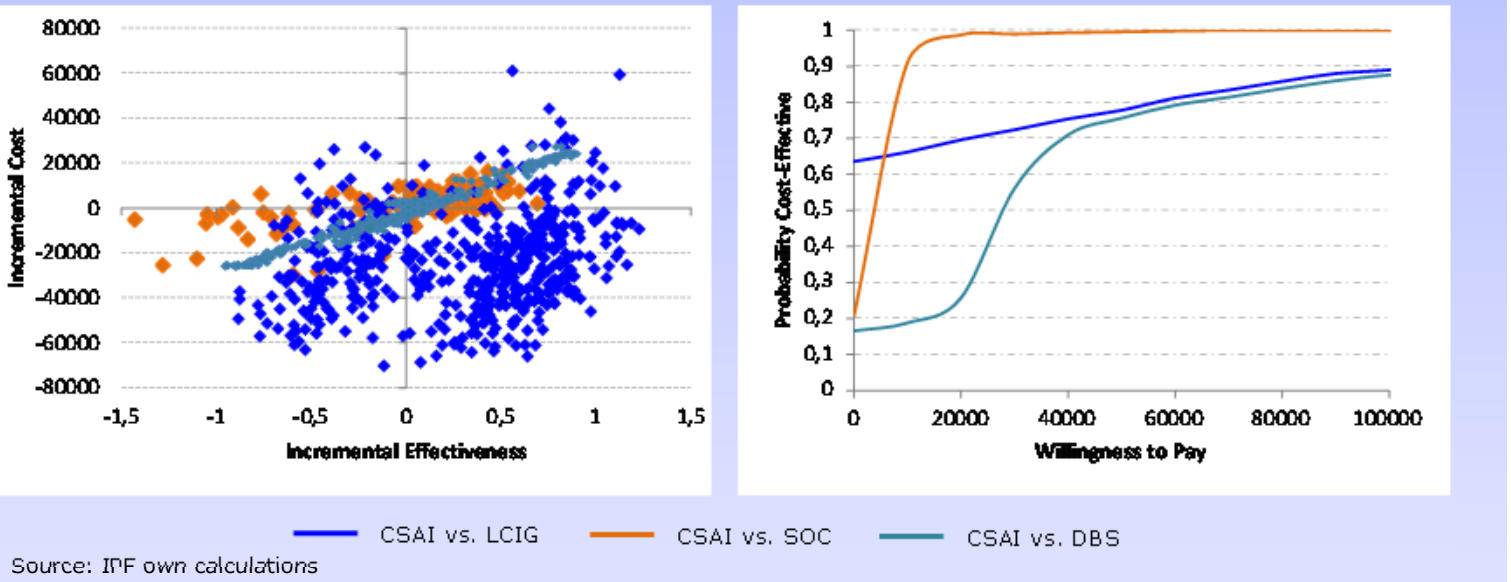
Incremental Cost
Incremental Effectiveness
Probability Cost-Effective
Willingness to Pay
CSAI vs. LCIG
CSAI vs. SOC
CSAI vs. DBS

Source: IPF own calculations

Monte Carlo probabilistic sensitivity analysis pools the results of 500 trials plotting incremental cost versus incremental effects. Based on this it creates acceptability curves to assess the likelihood of the comparators being cost-effective over a range of willingness to pay values in the UK setting. Sensitivity analysis revealed that CSAI is cost effective compared to LCIG in more than 69.6% (72.4%) of the simulations with a willingness to pay of 20,000 £ (30,000 £); versus DBS in 25.8% (56.2%) and versus SOC in 98.7% (98.9%) of all simulations (Fig. 3b). One-way sensitivity analysis shows the robustness of model.

Conclusion

Patients with advanced PD who are not eligible for alternate treatments are currently maintained on standard care and show a clear unmet need for alternative effective treatment options. The presented model has found a substantial incremental gain in QALYs for CSAI compared to SOC (0.23 QALYs) and to DBS (0.10 QALYs). CSAI could therefore be considered as cost-effective in the limited cohort of patients for whom it is indicated. The findings of this analysis must be considered in the light of the limitations outlined.
CSAI is a cost-effective alternative, reducing OFF-time and improving quality-of-life and is associated with a cost-advantage.

References

Lowin J, Bergman A, Ray Chaudhuri K et al. A cost-effectiveness analysis of levo-dopa/carbidopa intestinal gel compared to standard care in late stage Parkinson's disease in the UK. J Med Econ. 2011;14(5):584-93
Willis M, Persson U, Zoellner Y, et al. Reducing uncertainty in value-based pricing using evidence development agreements. The case of continuous intraduodenal infusion of levodopa/carbidopa (Duodopa) in Sweden. Applied Health Economics & Health Policy 2010;8:1-10
Findley LJ, Wood E, Lowin J, Roeder C, Bergman A, Schifflers M. The economic burden of advanced Parkinson's disease: an analysis of a UK patient dataset. J Med Econ. 2011;14(1):130-9
Findley LJ, Lees A, Apajasalo M, Pitkänen A, Turunen H. Cost-effectiveness of levodopa/carbidopa/entacapone (Stalevo) compared to standard care in UK Parkinson's disease patients with wearing-off. Curr Med Res Opin. 2005;21(7):1005–1014
Pollak P, Champay AS, Gaio JM, Hommel M, Benabid AL, Perret J. Subcutaneous administration of apomorphine in motor fluctuations in Parkinson's disease. Rev Neurol (Paris) 1990;146: 116–122
Gancher ST, Nutt JG, Woodward WR. Apomorphine infusional therapy in Parkinson's disease: clinical utility and lack of tolerance. Mov Disord 1995;10:37–43.
Katzenschlager R, Hughes A, Evans A, et al. Continuous subcutaneous apomorphine therapy improves dyskinesias in Parkinson's disease: a prospective study using single-dose challenges. Mov Disord 2005;20:151–157
Eggert K, Schrader C, Hahn M, et al. Continuous jejunal levodopa infusion in pa-tients with advanced parkinson disease: practical aspects and outcome of motor and non-motor complications. Clin Neuropharmacol 2008;31:151-66
Antonini A, Isaias IU, Canesi M, et al. Duodenal levodopa infusion for advanced Parkinson's disease: 12-month treatment outcome. Mov Disord 2007;22:1145-9
Deuschl G, Schade-Brittinger C, Krack P. A Randomized Trial of Deep-Brain Stimu-lation for Parkinson's Disease. N Engl J Med 2006;355:896-908
Additional Literature with the author

This study was funded by EVER Neuro Pharma Austria